SUPPORTING PATIENTS IN OPTIMAL PARTICIPATION AT WORK IN THE RHEUMATOLOGY CLINIC: RHEUMATOLOGISTS' AND NURSES' OPINION

Thu
0569-HPR

C.W.J.Logtenberg-Rutten RN [1], Y.J. van Eyk-Hustings H.C.S. R.N. [2], E.Stadermann [3], M.J.Scholte-Voshaar [4], J.M.W. Hazes M.D. Ph.D. [5], A.E.Boonen M.D. Ph.D.[2]

[1] Division of Clinical Immunology and Rheumatology, Academic Medical Center, Amsterdam, The Netherlands [2] Department of Rheumatology, Maastricht University Medical Center, Maastricht, The Netherlands
[3] Abbvie, Hoofddorp, The Netherlands [4,1] Patiënt Group Representive, The Netherlands, [5] Department of Rheumatology, Erasmus University Medical Center, Rotterdam, The Netherlands

TARGET@WORK

INTRODUCTION

Although participation in paid work is an important life area for patients and loss of work might have severe consequences for the physical, emotional and financial situation of patients, no recommendations for the assessment of work ability exist in rheumatology practice.

OBJECTIVE

The objective of this study was to explore whether rheumatologists and rheumatology nurses
a) Consider worker participation as a treatment target
b) Need tools to improve attention and care for participation in paid work.

METHODS

Study subjects:

Two comparable web-based surveys were developed and disseminated among the rheumatologists and rheumatology nurses in the Netherlands.

The questionnaires focussed on
a) Existing practice in assesment of workabillity of the patients
b) Priorities for different treatment targets (including work) and confidence with treatment plantreatms, to be scored on a 0/no priority- 10/highest priority scale
c) Need to have a standardized tool to adress problems at work
d) Possible barriers to apply such tool

RESULTS

a) 104 rheumatologists (40%) and 103 nurses (50%) filled in the survey.
b) 97% of the rheumatologists and 99 % of the nurses considered optimal worker participation a treatment target.
c) Both groups acknowledge the importance of workparticipation but the gap between priority and confidence with treatmentplans shows that support may be necessary.
d) Both rheumatologists and nurses considered a tool to assess problems at work and effective interventions important and necessary but effects should be examined.
e) Of the rheumatologists, 52 % limitations in time a barrier for an active approach in daily practice and 28% referred their patients with problems regarding workparticipation to the rheumatology nurse.

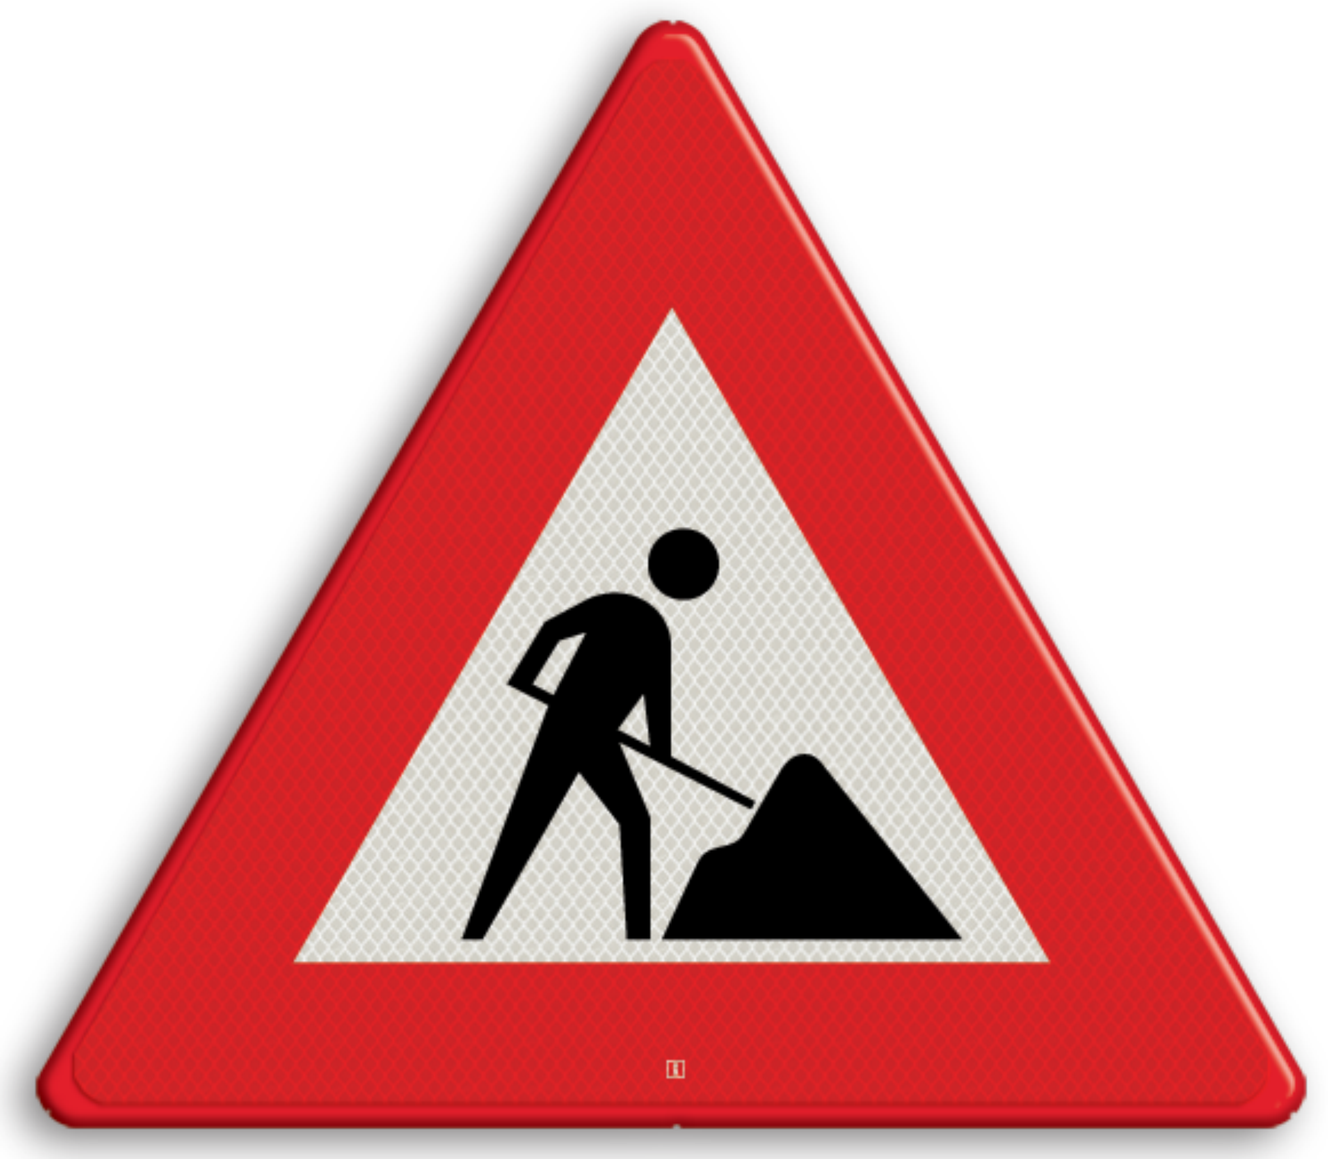

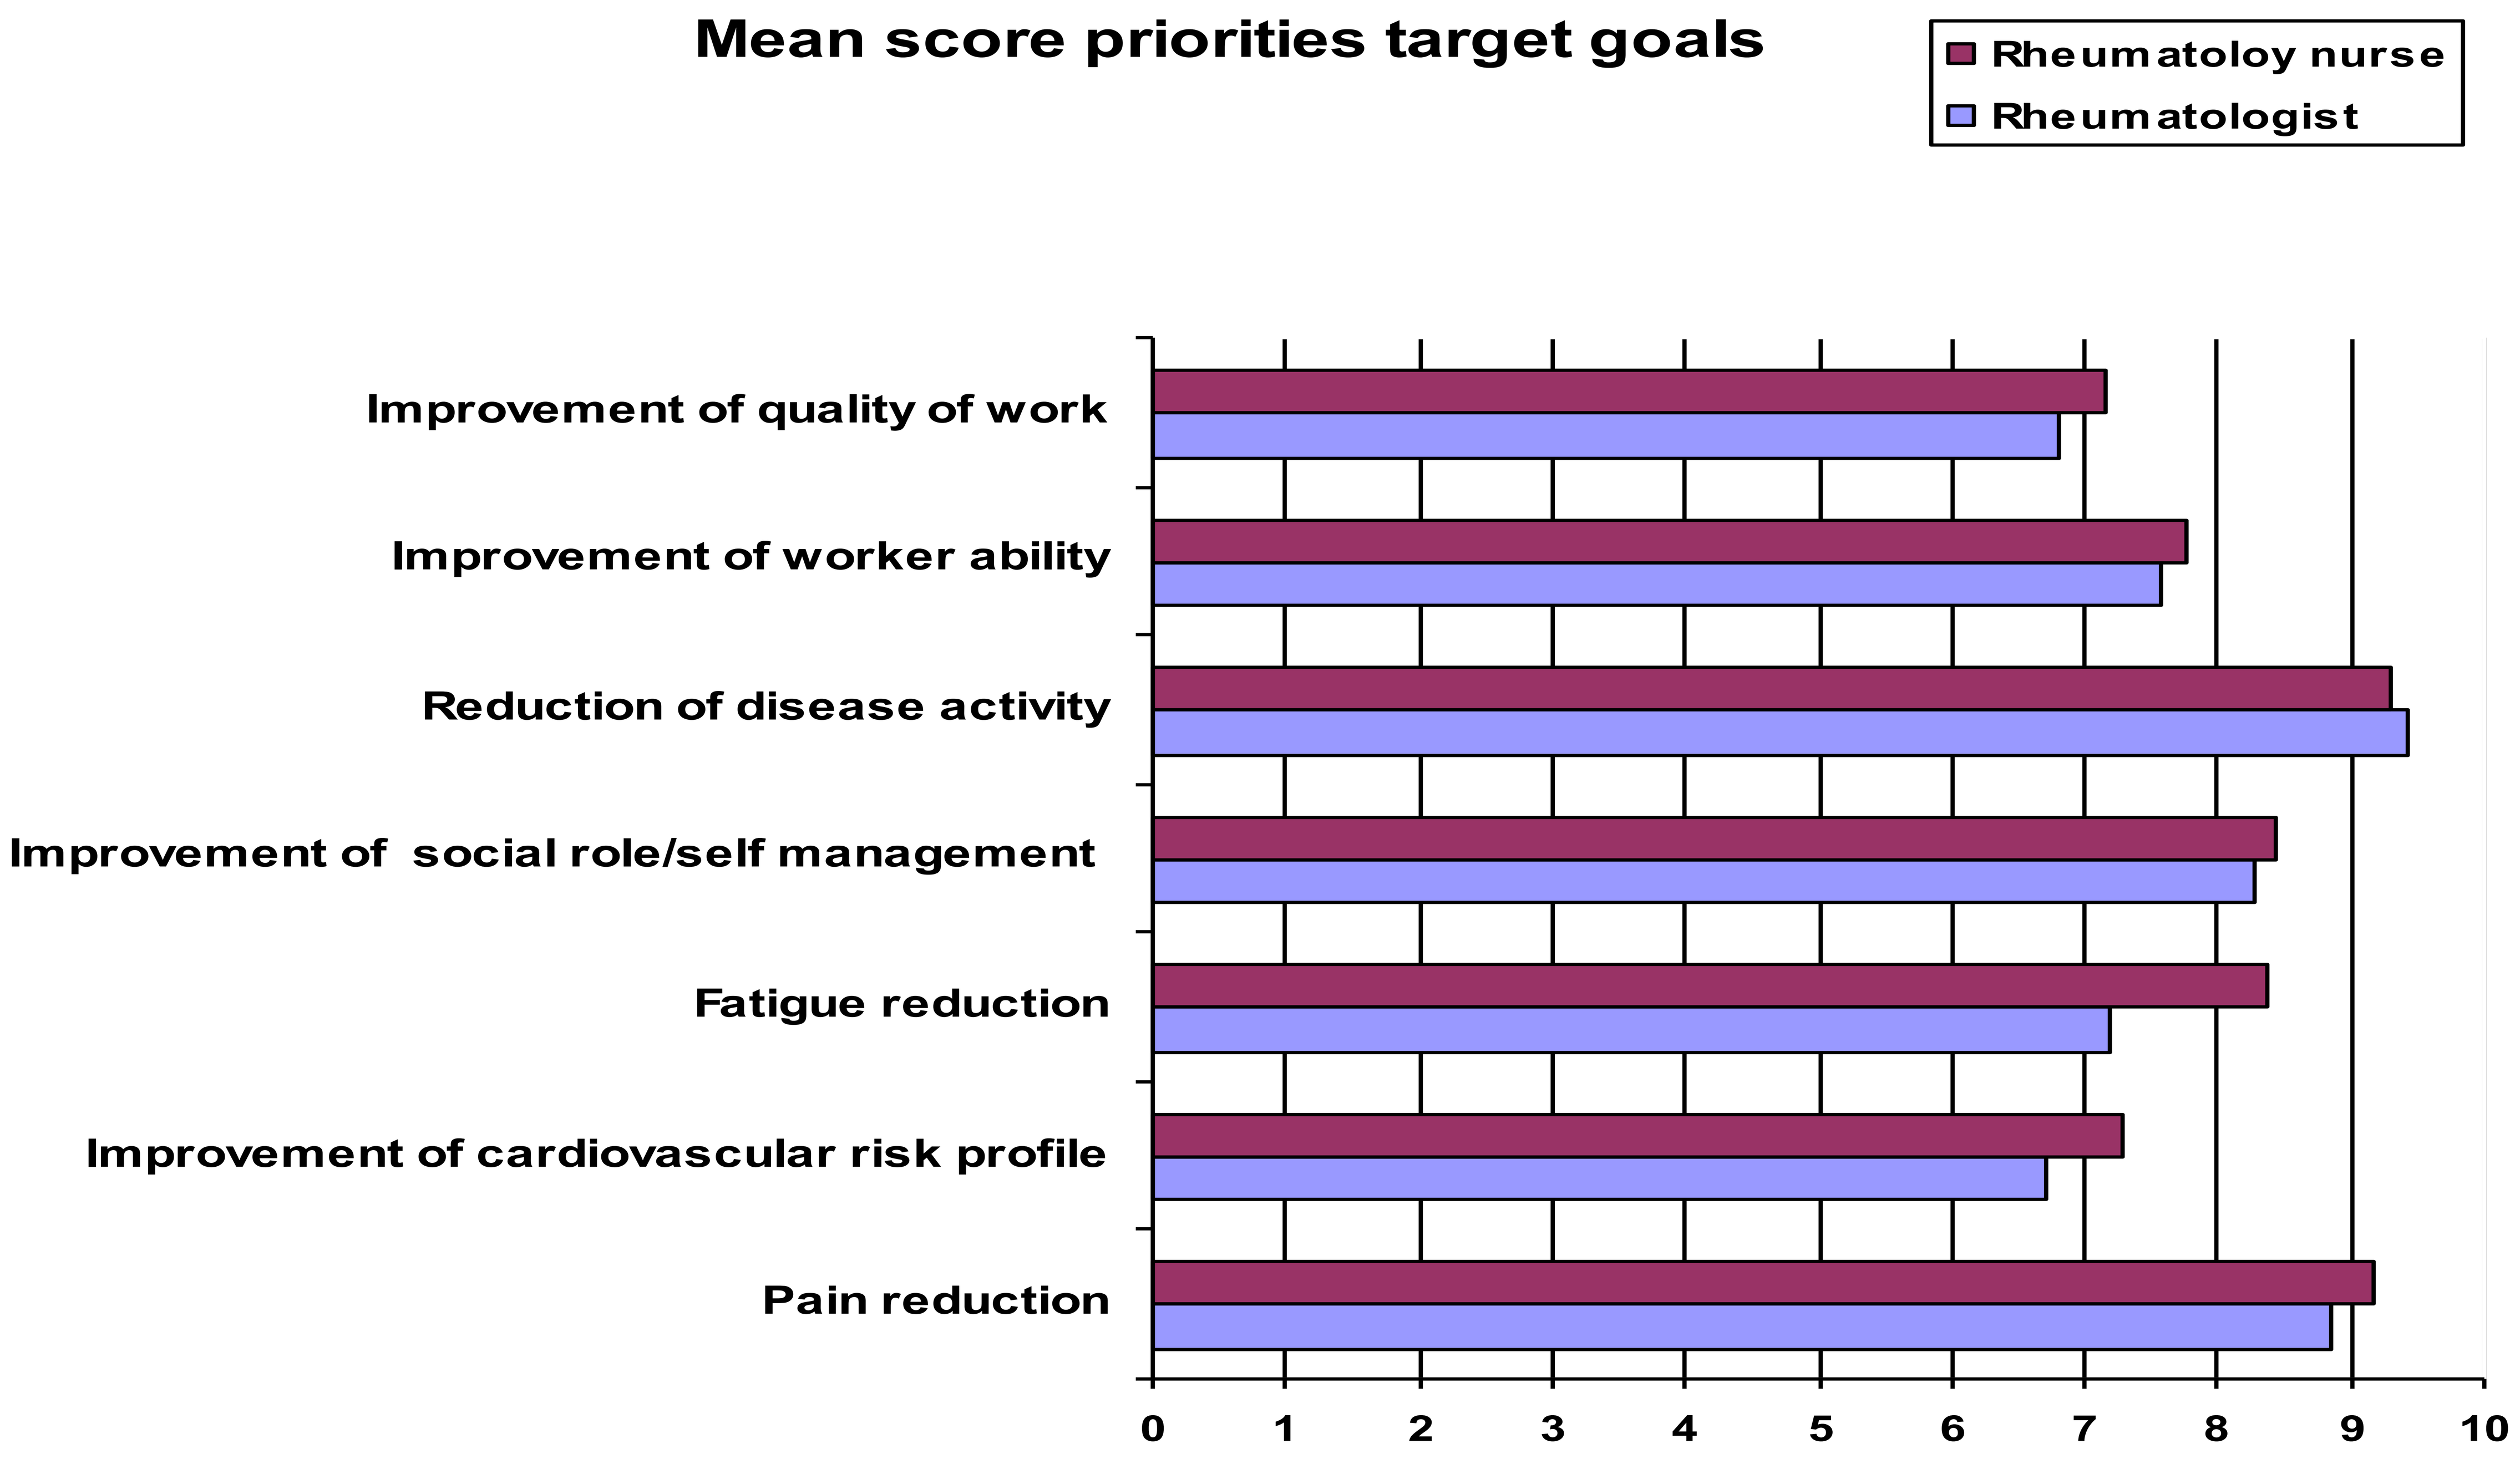
Mean score priorities target goals
Rheumatoloy nurse
Rheumatologist
Improvement of quality of work
Improvement of worker ability
Reduction of disease activity
Improvement of social role/self management
Fatigue reduction
Improvement of cardiovascular risk profile
Pain reduction
0
1
2
3
4
5
6
7
8
9
10

Priority treatment goal: Improve the quality of work

Functie			Frequentie	Cumulative Percent
Rhmtl	Valid	2	2	2.0
		3	3	5.0
		4	3	8.0
		5	13	21.0
		6	15	36.0
		7	28	64.0
		8	22	86.0
		9	10	96.0
		10	4	100.0
		Total	100	
Missing		System	4	
Total			104	
Nurse	Valid	0	1	1.0
		1	1	2.0
		2	2	4.0
		3	1	5.0
		4	3	7.9
		5	5	12.9
		6	11	23.8
		7	26	49.5
		8	36	85.1
		9	8	93.1
		10	7	100.0
		Total	101	
Missing		System	2	
Total			103	

CONCLUSION

The study shows that rheumatologists and nurses consider worker participation as an important treatment target. However, they express a need for (evidence based) tools to identify patients that are at risk for adverse work outcome and for better insight into effective interventions to improve the quality of care regarding work

Sponsored by abbvie

E-mail: c.w.logtenberg@amc.uva.nl